Interim Results of a Phase 1b/2 Study of ADG126 (a Masked anti-CTLA-4 SAFEbody®) Monotherapy and in Combination with Toripalimab (an anti-PD-1 Antibody) in Patients (pts) with Advanced / Metastatic Solid Tumors

ADAGENE

AACR 2023
Abstract Number: CT227

Mihitha Ariyapperuma[1], John J. Park[2], Adnan Khattak[3], Gary Richardson[4], Anis Hamid[4], Michelle Morris[5], Anthony W. Tolcher[6], Boon Cher Goh[7], Justina Lam[8], Bartosz Chmielowski[9], Kristine She[10], Yanyan Zhang[10], Ai Li[10], Songmao Zheng[10], Guizhong Liu[10], Lvyu Zhu[10], Hongyan Wang[10], Xiaoxing Cui[10], Peter Luo[10], Jiping Zha[10]*

*Presenting Author

1 One Clinical Research, Nedlands, WA, Australia; 2 Department of Clinical Medicine, Faculty of Medicine, Health and Human Sciences, Macquarie University, NSW, Australia; 3 One Clinical Research & Edith Cowan University, Perth, WA, Australia; 4 Cabrini Research, Malvern, VIC, Australia; 5 Sunshine Coast University Private Hospital, Birtinya, QLD, Australia; 6 NEXT Oncology, Texas, US; 7 National University of Singapore, Singapore; 8 National Cancer Centre, Singapore; 9 Jonsson Comprehensive Cancer Center at UCLA, Los Angeles, CA, USA; 10 Adagene Inc., San Diego, CA, USA .

Background

- ADG126 is a fully human anti-CTLA-4 IgG1 SAFEbody with a masking peptide that competes for antigen binding and subjects to cleavage in the tumor microenvironment (TME). This design enables ADG126 to be conditionally enriched and cleaved in the TME to provide a steady and prolonged drug exposure in the tumor while limiting peripheral circulation or off-tumor toxicity. Activated ADG126 binds to a unique and highly conserved epitope of CTLA-4 with species cross-reactivity for seamless translation studies
- ADG126 is designed to activate efficiently in vivo and prime T cells via partial blockade of CTLA-4 interaction with its ligands. Nonclinical studies have demonstrated that ADG126 depletes immunosuppressive Tregs through strong antibody-dependent cellular cytotoxicity (ADCC) specifically in the TME, and it synergizes with anti-PD-1 antibody to induce potent anti-tumor efficacy
- Plasma PK of total ADG126, intact ADG126, and cleaved ADG126* are approximately linear with dose. The half-life of total ADG126 is significantly higher than its parental antibody. Accumulation of cleaved ADG126 in plasma is observed at steady-state, likely contributed by ADG126 cleavage in TME

*Cleaved ADG126 was calculated by total ADG126 minus intact ADG126

Method

- This phase 1b/2, open-label, non-randomized study evaluates ADG126 monotherapy and ADG126 combination therapies in dose escalation, dose expansion and biopsy cohorts of pts with advanced / metastatic solid tumors (ADG126-1001, NCT04645069)
- Here we report interim data on the dose escalation, dose expansion and biopsy cohorts of ADG126 monotherapy as well as dose escalation of ADG126 + toripalimab (TORI)

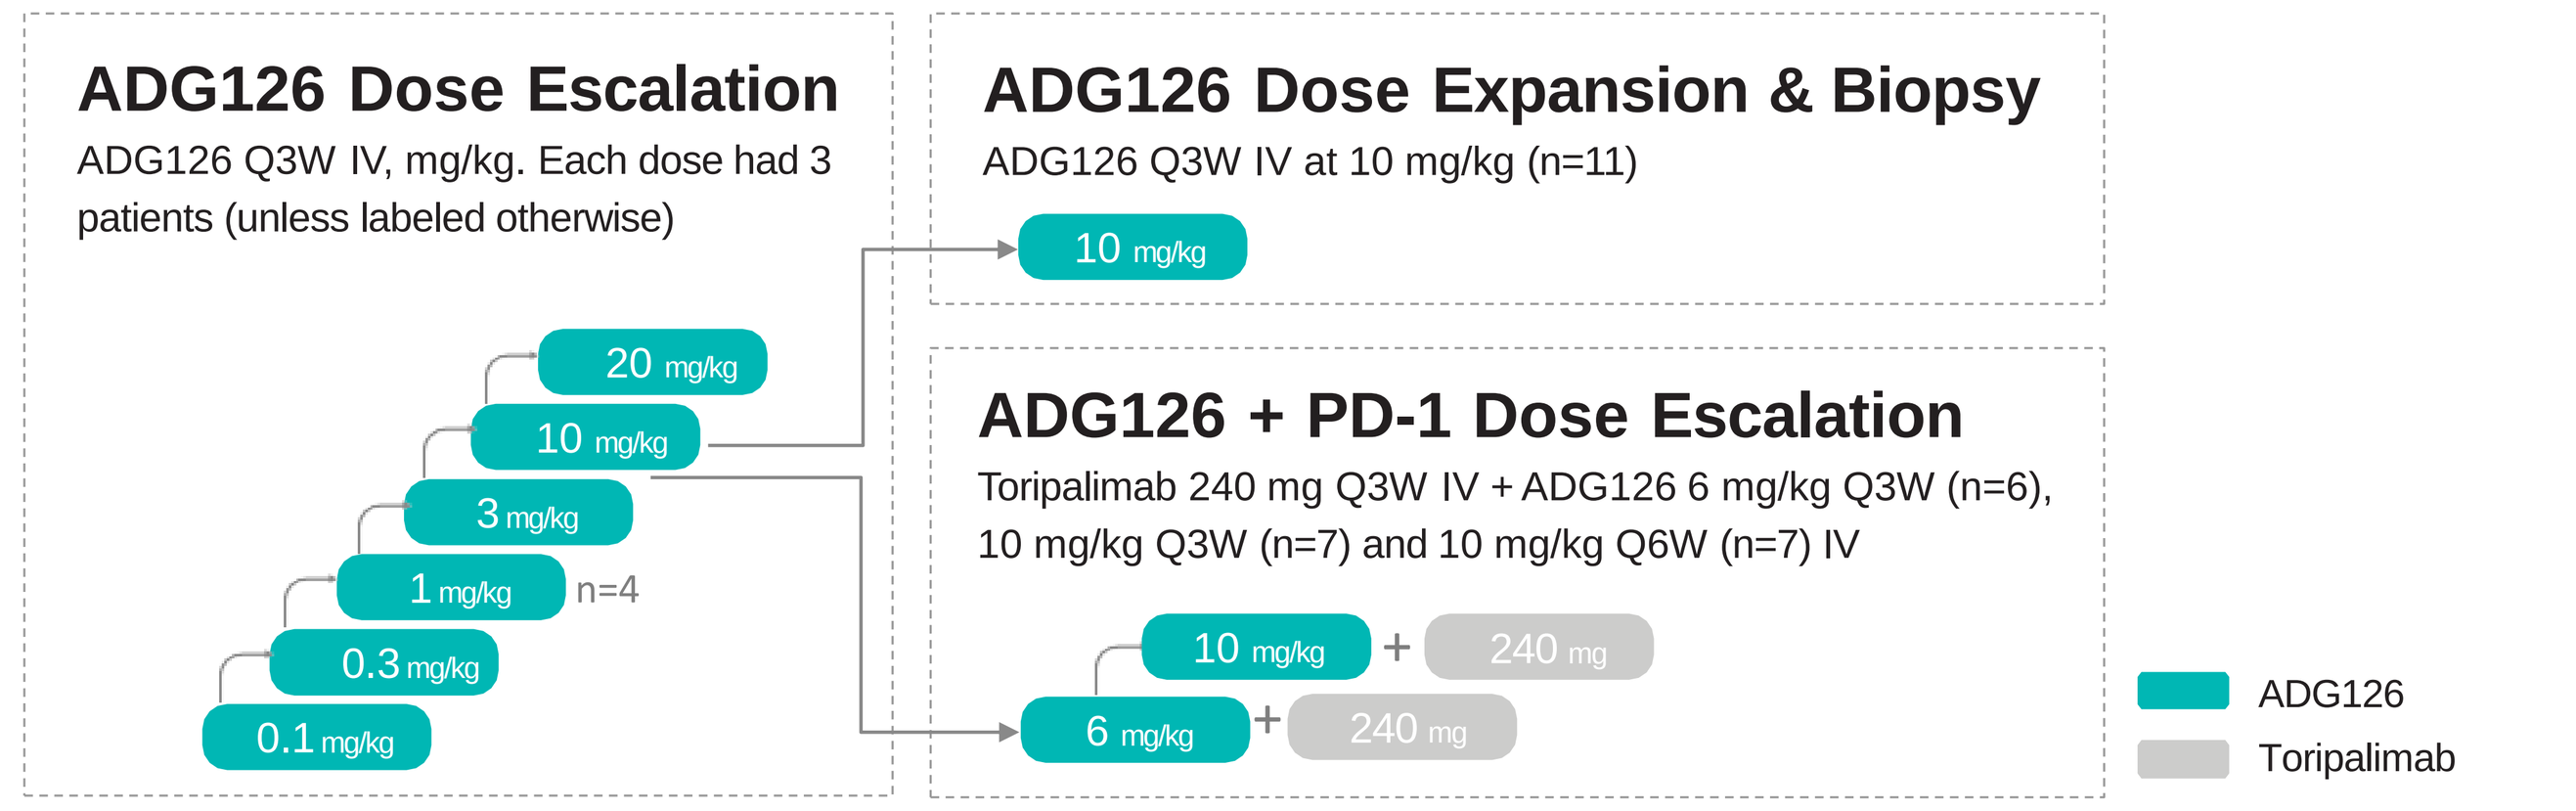
ADG126 Dose Escalation
ADG126 Q3W IV, mg/kg. Each dose had 3 patients (unless labeled otherwise)
20 mg/kg
10 mg/kg
3 mg/kg
1 mg/kg n=4
0.3 mg/kg
0.1 mg/kg
ADG126 Dose Expansion & Biopsy
ADG126 Q3W IV at 10 mg/kg (n=11)
10 mg/kg
ADG126 + PD-1 Dose Escalation
Toripalimab 240 mg Q3W IV + ADG126 6 mg/kg Q3W (n=6), 10 mg/kg Q3W (n=7) and 10 mg/kg Q6W (n=7) IV
10 mg/kg + 240 mg
6 mg/kg + 240 mg
ADG126
Toripalimab

Primary endpoints: Safety and tolerability; determine maximum administered dose, maximum tolerated dose and recommended Phase 2 doses. Secondary endpoints: PK, anti-drug antibodies (ADA), ORR, DCR, DOR, PFS and OS. Key inclusion criteria: Patients with advanced or metastatic solid tumors with ECOG ≤ 1 and at least one measurable lesion per RECIST1.1. Imaging was performed every 6 weeks for the first 4 cycles, then every 9 weeks afterwards. Tumor response was investigator-determined using RECIST 1.1 and iRECIST. Data cutoff: March 14, 2023; n = number of patients by data cutoff date; IV = intravenous; Q3W = Every 3 weeks; Q6W = Every 6 weeks

Patient Demographics, Characteristics & Drug Exposure

- Across all dose levels, 30 pts received ADG126 monotherapy
- Twenty pts received ADG126 (6 mg/kg Q3W and 10 mg/kg Q3W or Q6W) + TORI (240 mg Q3W)
- Pts were heavily pre-treated
- Over 15 tumor types were included in the study, including "cold tumors" or immune-experienced "hot tumors". The most common tumor types include colorectal cancer and ovarian cancer

*Study treatment cycles includes ADG126 and/or TORI

Table 1. Patient Demographics, Characteristics and Drug Exposure

Characteristics	ADG126 (N = 30)	ADG126 + TORI (N = 20)
Age (years), median (range)	63.5 (39, 84)	59.5 (36, 85)
Female, n (%)	16 (53)	10 (50)
Race, n (%)		
Caucasian, n (%)	27 (90)	15 (75)
Asian, n (%)	3 (10)	4 (20)
Native American, n (%)	0	0
Aboriginal Australian, n (%)	0	1 (5)
ECOG PS, n (%)		
0	16 (53)	9 (45)
1	14 (47)	11 (55)
Number of prior lines of treatment, n (%)		
≥3	16 (53)	11 (55)
Prior immunotherapy, n (%)	14 (47)	7 (35)
Study treatment cycles		
Median (range)	2 (1, 25)	4 (1, 10)*
≥ 4 cycles, n (%)	9 (30)	13 (65)*

Conclusion

- ADG126 (anti-CTLA-4 SAFEbody) monotherapy is well tolerated up to 20mg/kg Q3W and demonstrated promising efficacy signals in heavily pre-treated patients
 - Prolonged stable disease in 5 patients
 - Increased ratio of Teff / Treg, with Treg depletion and increased CD8+ T cells observed in paired tumor biopsies
- The safety profile of ADG126 + TORI dose escalation demonstrates best-in-class potential in comparison with other anti-CTLA-4 molecules in combination with anti-PD-1 antibody at the similar doses / schedules
- ADG126 + TORI shows encouraging efficacy, including two confirmed PRs, as well as prolonged stable disease and reduced target lesions in "cold" GI tumors
 - ORR = 28% and DCR = 57% among the 7 evaluable patients who received ADG126 10 mg/kg Q3W + TORI 240 mg Q3W
 - Prolonged stable disease as well as a 58% and 21% reduction in the sum of target lesion was observed in two MSS CRC patients with liver metastasis at baseline; similar finding was also observed in a PDAC patient with a 5% reduction in the sum of target lesions
- Continuous dosing beyond 4 cycles enabled by the favorable safety profile may enable combination with agents beyond anti-PD-1 therapy: a study of ADG126 + atezolizumab + bevacizumab (NCT04524871) is being planned
- The unique mechanism of Treg depletion may unleash the potential of ADG126 in addressing significant unmet need in "cold tumors". Dose expansion of ADG126 10 mg/kg + TORI is planned in indications including MSS CRC

Safety: ADG126 Monotherapy

- Well tolerated with no dose limiting toxicities at doses up to 20 mg/kg Q3W with repeat dosing
- No > G2 treatment-related adverse events (TRAEs) and no treatment-related serious adverse events (SAEs)
- No patient discontinued study treatment due to TRAE

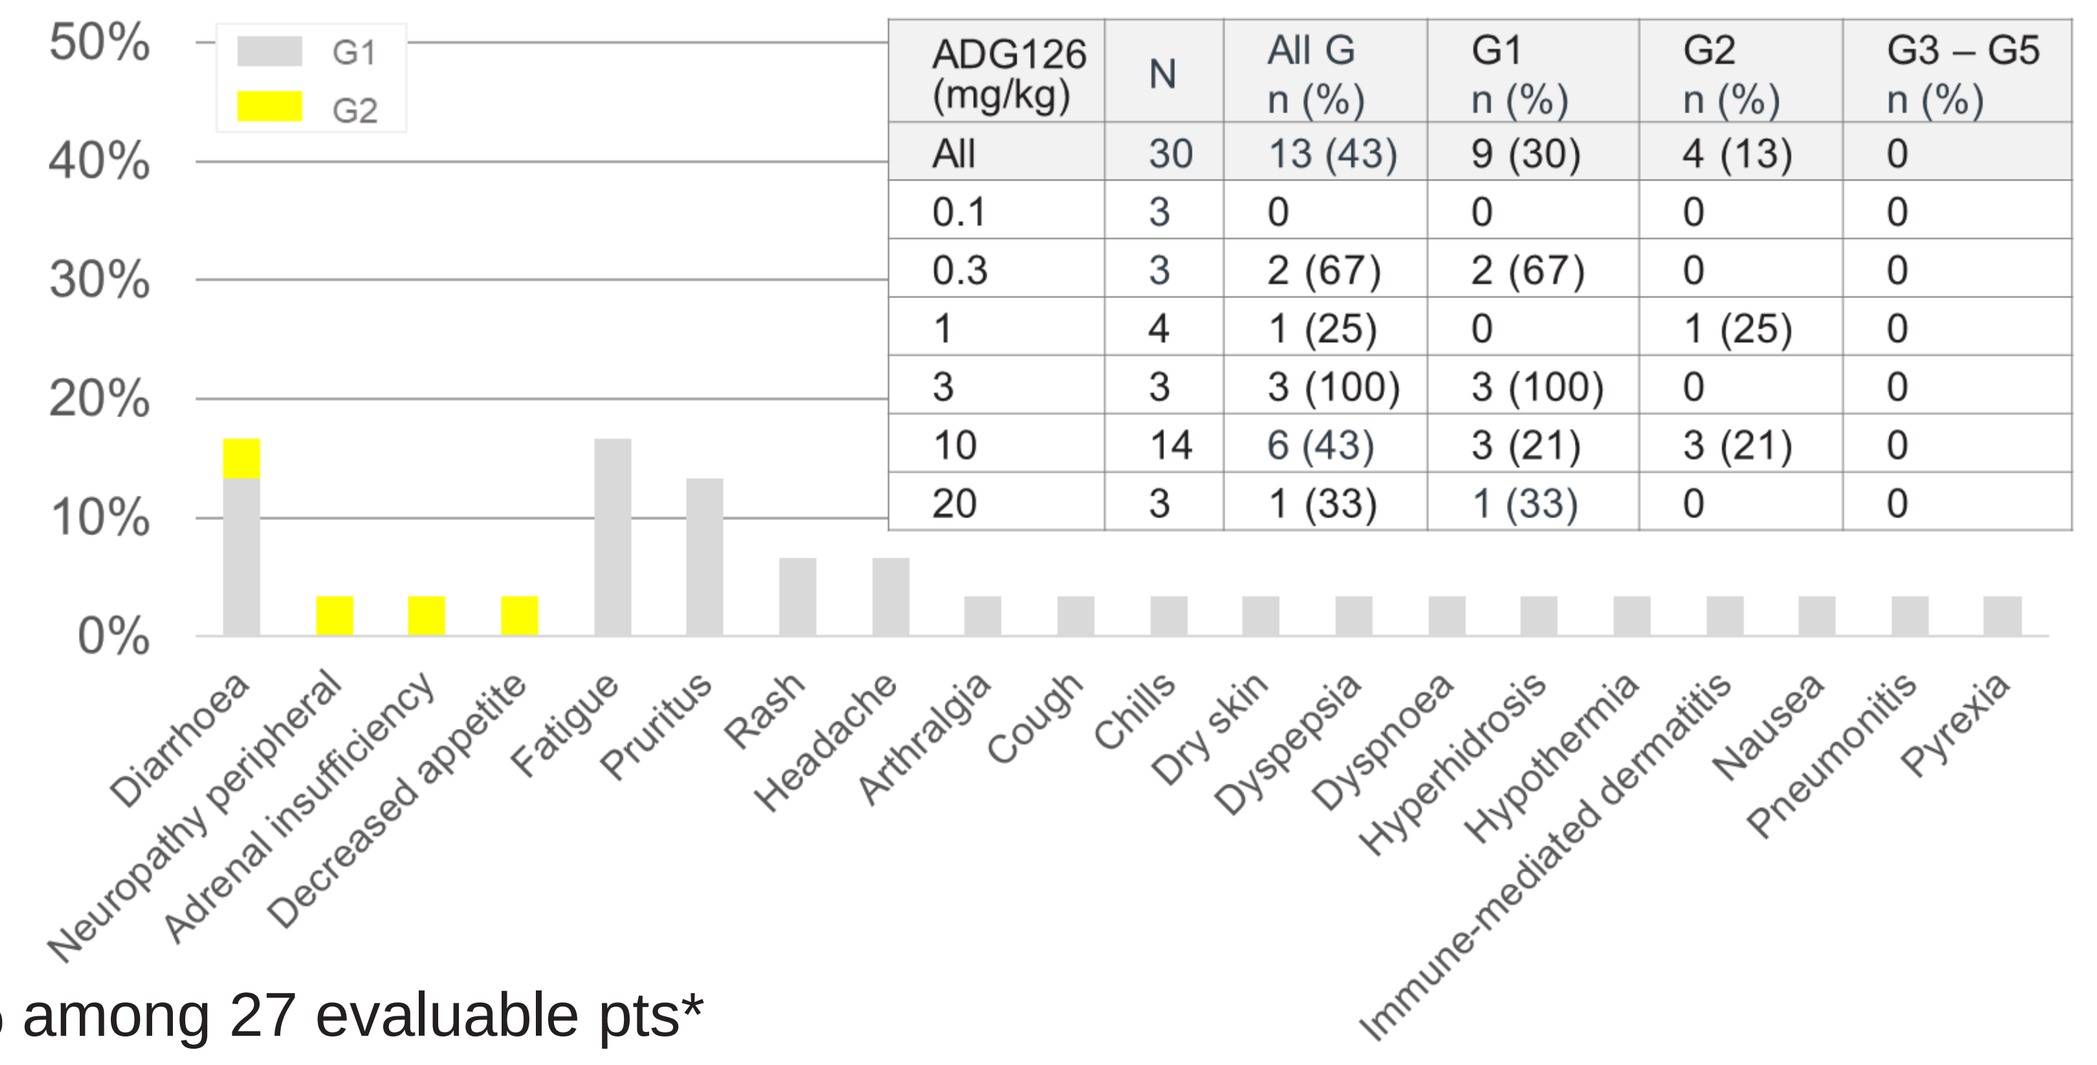
50%
40%
30%
20%
10%
0%
G1
G2
Diarrhoea
Neuropathy peripheral
Adrenal insufficiency
Decreased appetite
Fatigue
Pruritus
Rash
Headache
Arthralgia
Cough
Chills
Dry skin
Dyspepsia
Dyspnoea
Hyperhidrosis
Hypothermia
Immune-mediated dermatitis
Nausea
Pneumonitis
Pyrexia

ADG126 (mg/kg)	N	All G n (%)	G1 n (%)	G2 n (%)	G3 – G5 n (%)
All	30	13 (43)	9 (30)	4 (13)	0
0.1	3	0	0	0	0
0.3	3	2 (67)	2 (67)	0	0
1	4	1 (25)	0	1 (25)	0
3	3	3 (100)	3 (100)	0	0
10	14	6 (43)	3 (21)	3 (21)	0
20	3	1 (33)	1 (33)	0	0

Figure 2 and Table 2. TRAEs reported at any dose level of ADG126 monotherapy

Efficacy: ADG126 Monotherapy

- Across all dose levels, disease control rate (DCR) = 37% among 27 evaluable pts*
- Prolonged stable disease was observed in 5 pts, including 14 cycles for a NSCLC pt and 22 cycles for an ovarian carcinoma pt
 - The ovarian serous carcinoma pt experienced SD (ADG126 1 mg/kg Q3W) with a 90% reduction in CA125 and a 22% reduction in the sum of target lesions
 - A 12% reduction in the sum of target lesions was observed in the NSCLC pt who previously progressed on pembrolizumab and docetaxel; this pt completed 14 cycles of ADG126 20 mg/kg Q3W with no TRAEs

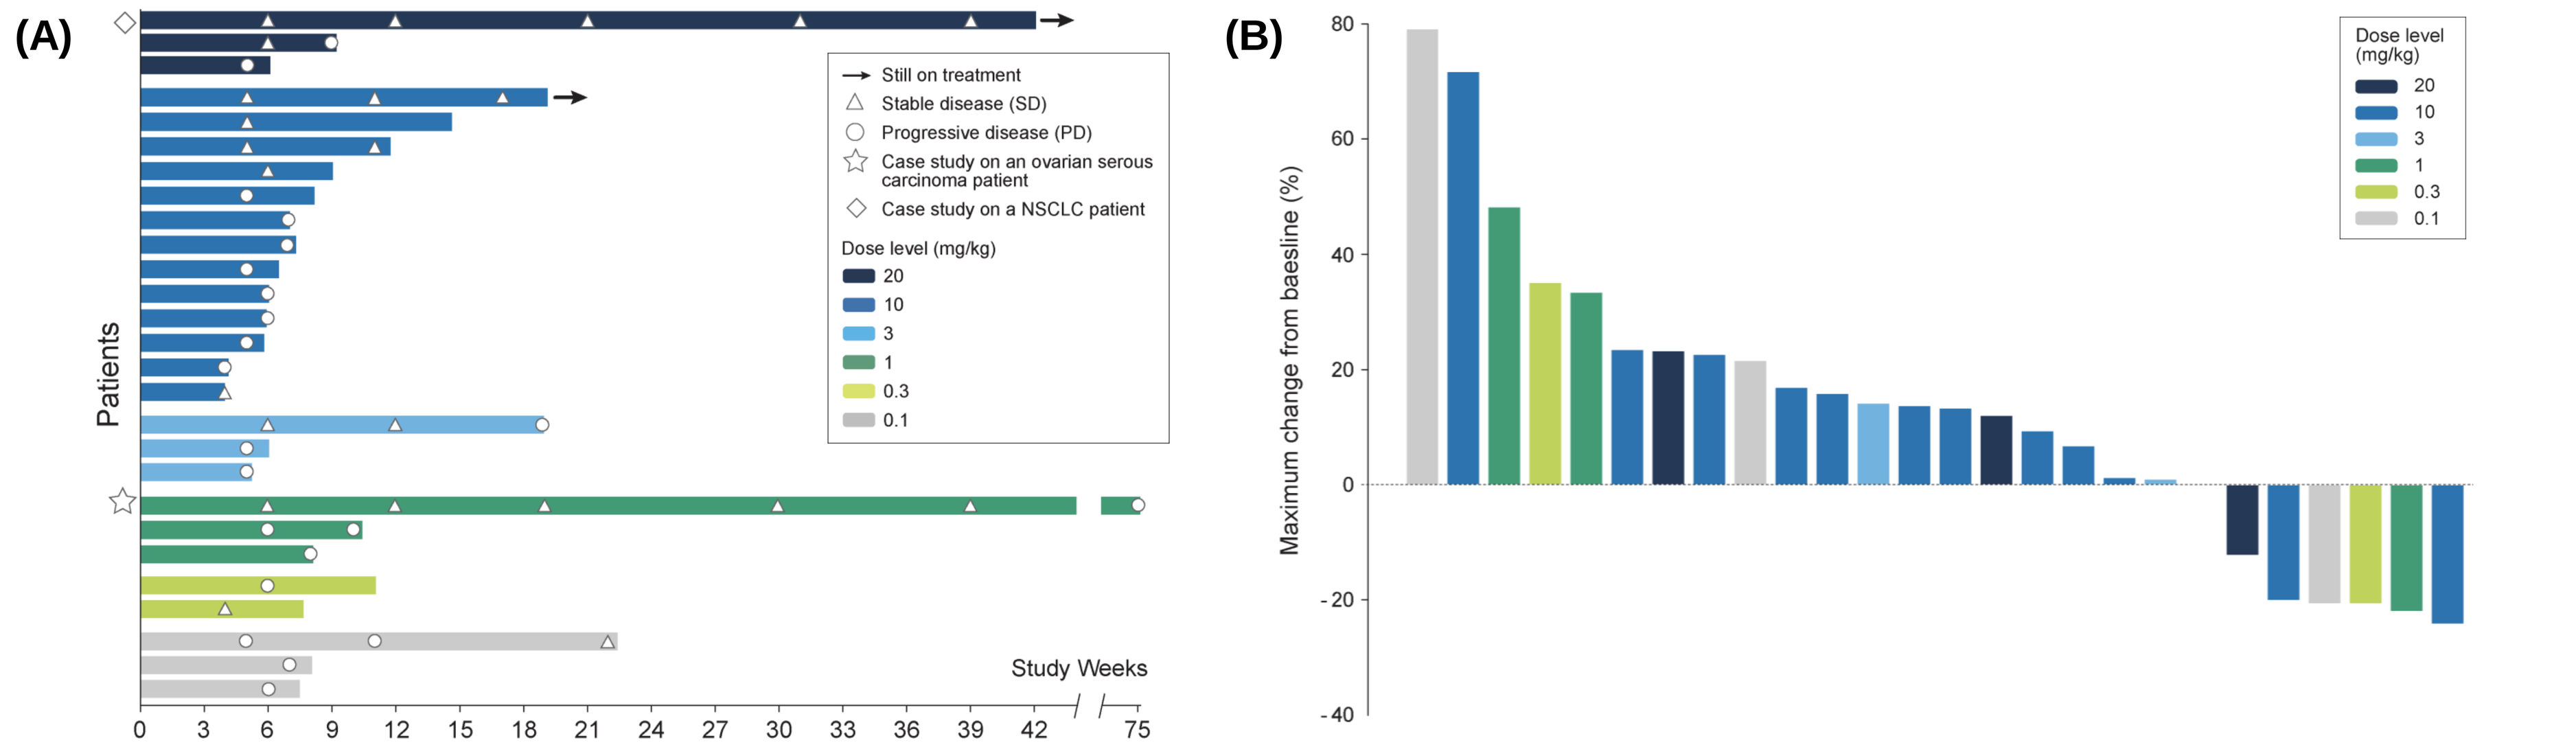
(A)
Still on treatment
Stable disease (SD)
Progressive disease (PD)
Case study on an ovarian serous carcinoma patient
Case study on a NSCLC patient
Dose level (mg/kg)
20
10
3
1
0.3
0.1
Patients
Study Weeks
0 3 6 9 12 15 18 21 24 27 30 33 36 39 42 75
(B)
Maximum change from baseline (%)
Dose level (mg/kg)

Figure 3. Response to ADG126 monotherapy in 27 evaluable pts* (A) swimmer plot (B) waterfall plot of response. *Evaluable pts with at least one valid post-baseline tumor assessment. One PD pt did not have all target lesions measured at post-baseline and was not included in the waterfall plot. For swimmer plot, bars end at study day of last dose, end of treatment (EOT) date (if EOT date is available) or last tumor assessment date, whichever is latest; three tumor assessments (SD) were truncated from the bar of the ovarian serous carcinoma patient due to spacing

Case study: Increased Teff / Treg with Treg Depletion in TME of an HCC Patient who Progressed on Atezolizumab + Bevacizumab

- Male, 39 y, ECOG PS 0, stage IIIB hepatocellular carcinoma
- Previously progressed on anti-PD-L1 therapy
 - 1L: Atezolizumab + bevacizumab (Jul 2021 – Feb 2022, PD)
 - 2L: Lenvatinib (Mar – Sep 2022, PD)
- Ongoing ADG126 10 mg/kg Q3W in Cycle 6 with control of tumor growth (stable disease)
- Increased ratio of Teff / Treg, with Treg depletion and increased CD8+ T cells was observed in paired tumor biopsies

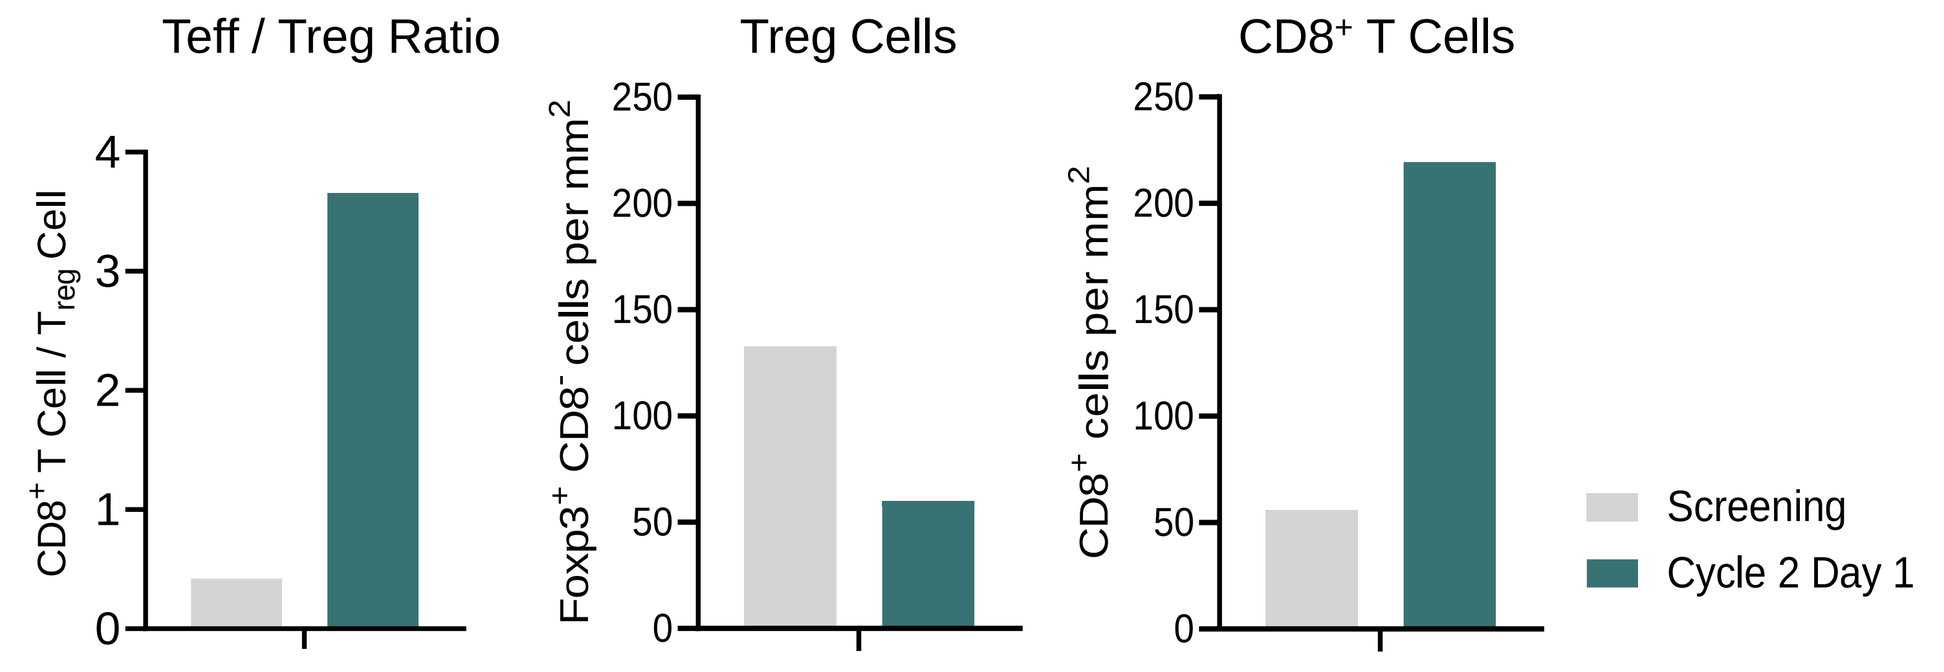
Teff / Treg Ratio
Treg Cells
CD8+ T Cells
CD8+ T Cell / Treg Cell
Foxp3+ CD8- cells per mm2
CD8+ cells per mm2
Screening
Cycle 2 Day 1

Figure 4. CD8 (green) / Foxp3 (red) / Pan-CK (yellow) / DAPI (blue) immunofluorescence of FFPE tumor biopsy samples. Paired tumor biopsies were collected before and after treatment. Multiplex immunofluorescence analysis was performed by Dr. Joe Yeong's lab at IMCB, A*STAR. Images were analyzed using HALO. Tregs were defined as Foxp3+ CD8- cells. Teff cells were defined as CD8+ T cells

Safety: ADG126 + Toripalimab Dose Escalation

- No significant differences in safety across the three dose escalation cohorts
 - No DLT or >G3 TRAE has been reported
 - Five pts (25%) experienced G3 TRAEs: Most of G3 TRAEs occurred no earlier than Cycle 4
 - Three pts experienced treatment-related SAEs, including one G3 hepatitis§, one G3 sepsis‡ and one G3 myocarditis‡ (this led to discontinuation of study)

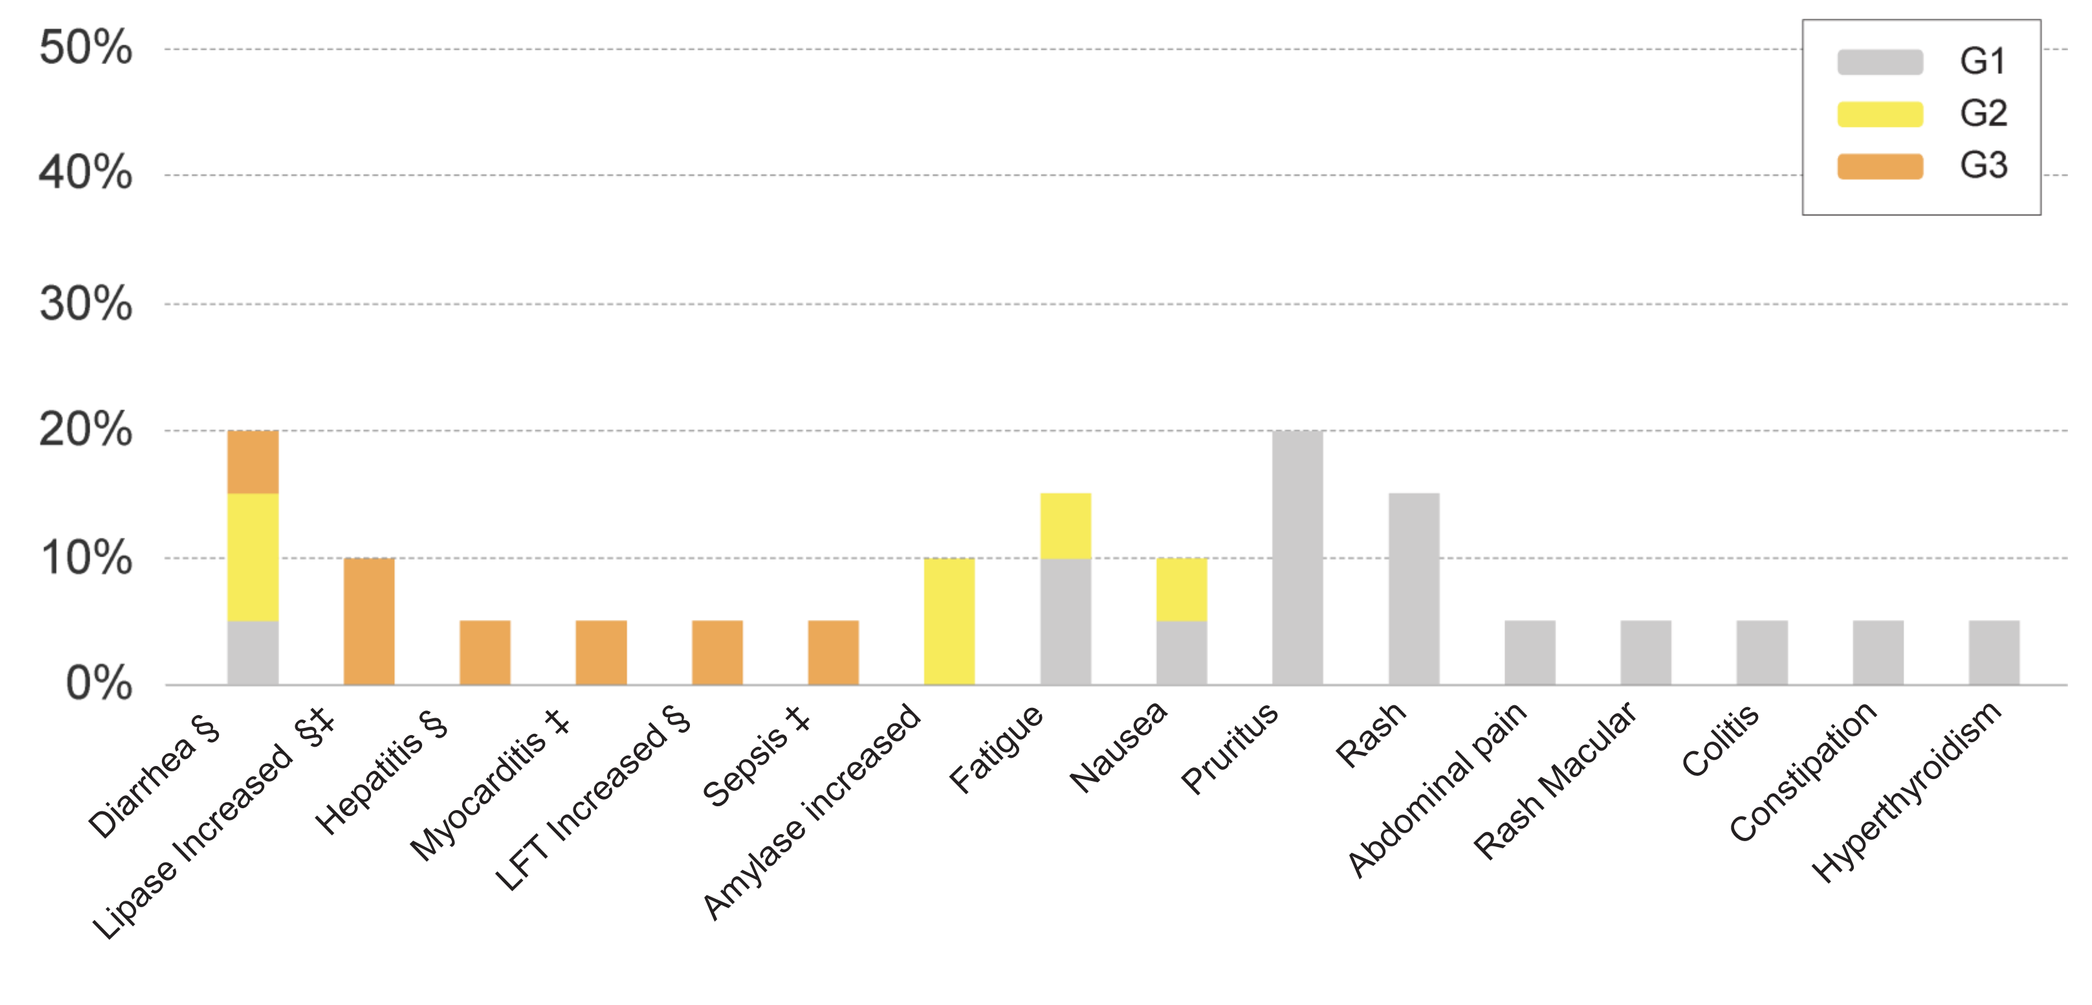
G1
G2
G3
Diarrhea §
Lipase Increased §‡
Hepatitis §
Myocarditis ‡
LFT Increased §
Sepsis ‡
Amylase increased
Fatigue
Nausea
Pruritus
Rash
Abdominal pain
Rash Macular
Colitis
Constipation
Hyperthyroidism

Figure 5. TRAEs in 20 pts of three dose escalation cohorts who received ADG126 (6 mg/kg Q3W and 10 mg/kg Q3W or Q6W) + TORI (240mg Q3W). LFT = Liver function test; § G3 TRAE observed at ADG126 6 mg/kg Q3W + TORI 240 mg Q3W; ‡ G3 TRAE observed at ADG126 10 mg/kg Q3W or Q6W + TORI 240 mg Q3W

Efficacy: ADG126 + Toripalimab Dose Escalation

- Among the 18 evaluable pts* in the three dose escalation cohorts who received ADG126 (6 mg/kg Q3W and 10 mg/kg Q3W or Q6W) + TORI (240 mg Q3W), overall ORR = 11% and DCR = 56%
 - ORR = 28% and DCR = 57% among the 7 evaluable pts* who received ADG126 10 mg/kg Q3W + TORI 240 mg Q3W
 - Confirmed PR observed in two pts with penile SCC and anal SCC who were previously treated with chemotherapy
- Prolonged stable disease was observed in a PDAC pt with a 5% reduction in the sum of target lesions
- A 58% and 21% reduction in the sum of target lesions were observed in two MSS CRC pts with liver metastasis at baseline, respectively

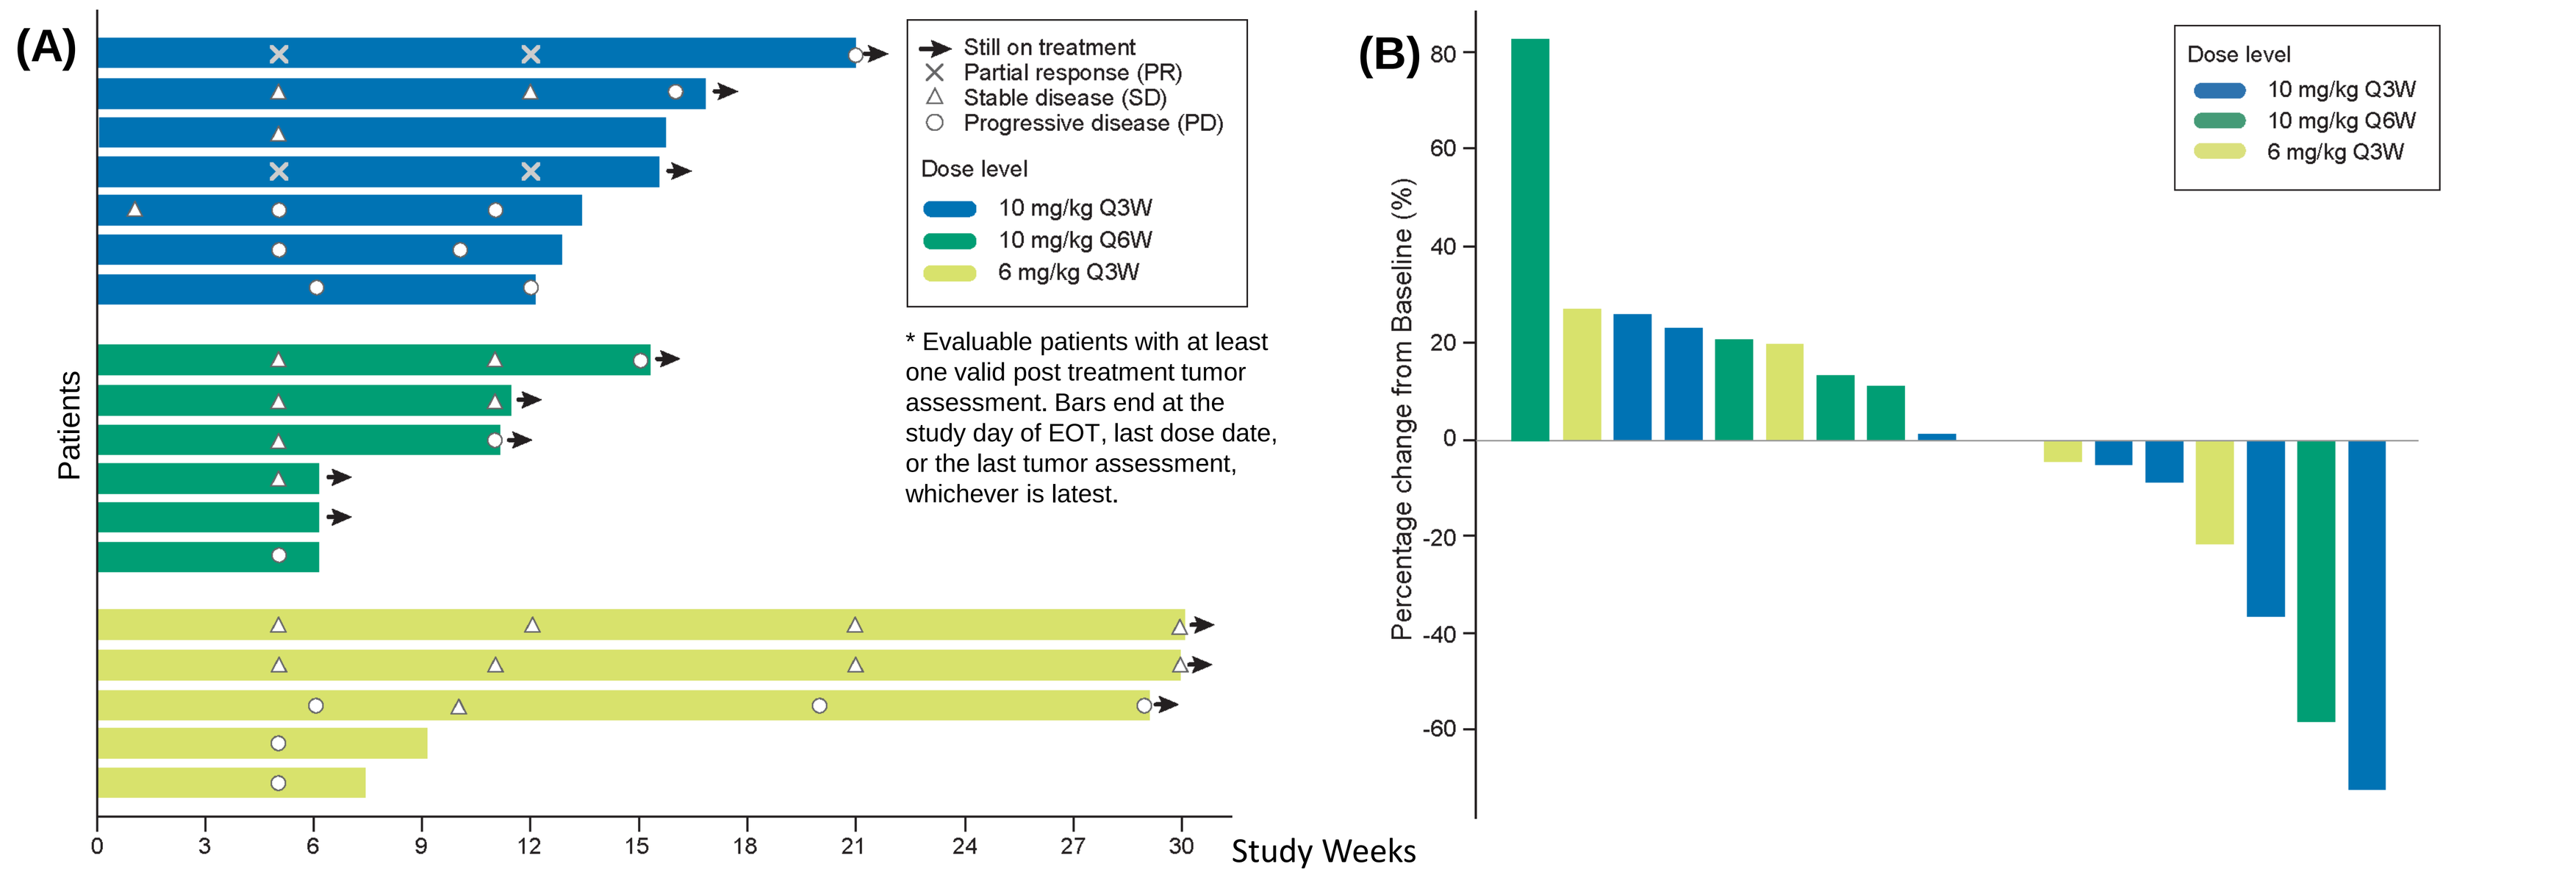
(A)
Still on treatment
Partial response (PR)
Stable disease (SD)
Progressive disease (PD)
Dose level
10 mg/kg Q3W
10 mg/kg Q6W
6 mg/kg Q3W
* Evaluable patients with at least one valid post treatment tumor assessment. Bars end at the study day of EOT, last dose date, or the last tumor assessment, whichever is latest.
Patients
Study Weeks
(B)
Percentage change from Baseline (%)
Dose level

Figure 6. Response to ADG126 in combination with toripalimab in 18 evaluable pts* of three dose escalation cohorts who received ADG126 (6 mg/kg Q3W and 10 mg/kg Q3W or Q6W) + TORI (240mg Q3W) (A) swimmer plot (B) waterfall plot

Case Studies: Prolonged Stable Disease and Reduced Target Lesions in "Cold" Gastrointestinal Epithelial Tumors

- Male, 53 y, ECOG PS 0
- MSS rectosigmoid adenocarcinoma with liver metastasis at baseline
- Previously received curative surgery and palliative surgery for liver metastasis and 3 lines of therapies: Leucovorin + 5FU + irinotecan + oxaliplatin, bevacizumab + TAS102 and regorafenib
- Mixed response with a 58% reduction in the sum of target lesions was observed at week 17; treatment is ongoing (ADG126 10 mg/kg Q6W + TORI 240 mg Q3W)

		Baseline	Week 7	Week 13	Week 17
Target Lesion	TL1 – Lung	15 mm	14 mm	13 mm	16 mm
	TL2 – Lymph Node	22 mm	12 mm	12 mm	Disappeared
	TL3 – Lymph Node	18 mm	10 mm	6 mm	Disappeared
	TL4 – Lymph Node	19 mm	14 mm	15 mm	Disappeared
	TL5 – Liver	17 mm	20 mm	22 mm	22 mm
	Sum	91 mm	70 mm (-23%)	68 mm (-25%)	38 mm (-58%)
Non-Target Lesion	NTL1 – Lung	Present	Present	Present	Disappeared
	NTL2 – Bone	Present	Present	Present	Present
New Lesion		NA	No	No	Yes
Overall Response		NA	SD	SD	PD (iuPD)

- Male, 74 y, ECOG PS 1, MSS colorectal adenocarcinoma with liver metastasis at baseline. Previously received 3 lines of therapies: XELOX + bevacizumab; irinotecan + cetuximab; irinotecan + panitumumab
- A 21% reduction in the sum of target lesions was observed; treatment is ongoing (ADG126 6 mg/kg Q3W + TORI 240 mg Q3W)

		Baseline	Week 8	Week 14	Week 24	Week 33
Target Lesion	TL1 - Liver	55 mm	52 mm	44 mm	40 mm	38 mm
	TL2 - Liver	48 mm	48 mm	44 mm	44 mm	43 mm
	Sum	103 mm	100 mm (-3%)	88 mm (-15%)	84 mm (-18%)	81 mm (-21%)
Non-Target Lesion		Present	Present	Present	Present	Present
New Lesion		NA	No	No	No	No
Overall Response		NA	SD	SD	SD	SD

- Female, 56 y, ECOG PS 0, pancreatic ductal adenocarcinoma. Previously received curative surgery and 4 lines of therapies: gemcitabine / nab-paclitaxel; FOLFIRINOX; oxaliplatin, raltitrexed, irinotecan and leucovorin; gemcitabine / capecitabin
- Prolonged control of tumor growth (SD) with a 5% reduction in the sum of target lesions; treatment is ongoing (ADG126 6 mg/kg Q3W + TORI 240 mg Q3W)

		Baseline	Week 8	Week 15	Week 23	Week 31
Target Lesion	TL1 - Liver	23 mm	22 mm	23 mm	23 mm	17 mm
	TL2 - Liver	21 mm	21 mm	20 mm	19 mm	25 mm
	Sum	44 mm	43 mm (-2%)	43 mm (-2%)	42 mm (-5%)	42 mm (-5%)
Non-Target Lesion		Present	Present	Present	Present	Present
New Lesion		NA	No	No	No	No
Overall Response		NA	SD	SD	SD	SD

Confirmed PRs in Two Pts

- Female, 51 y, ECOG PS 0, metastatic anal squamous cell carcinoma
- Previously received fluorouracil (5FU) / mitomycin C + radiation
- ADG126 10 mg/kg Q3W + TORI 240 mg Q3W (ongoing in Cycle 5)
- Confirmed PR with a 36% reduction in the sum of target lesions

- Male, 64 y, ECOG PS 1, metastatic penile SCC
- Previously received partial penectomy, bilateral lymph node dissection, chemotherapy (paclitaxel, ifosfamide, cisplatin) and palliative RT
- ADG126 10 mg/kg Q3W + TORI 240 mg Q3W (ongoing in Cycle 6)
- Confirmed PR with a 72% reduction in the sum of target lesions

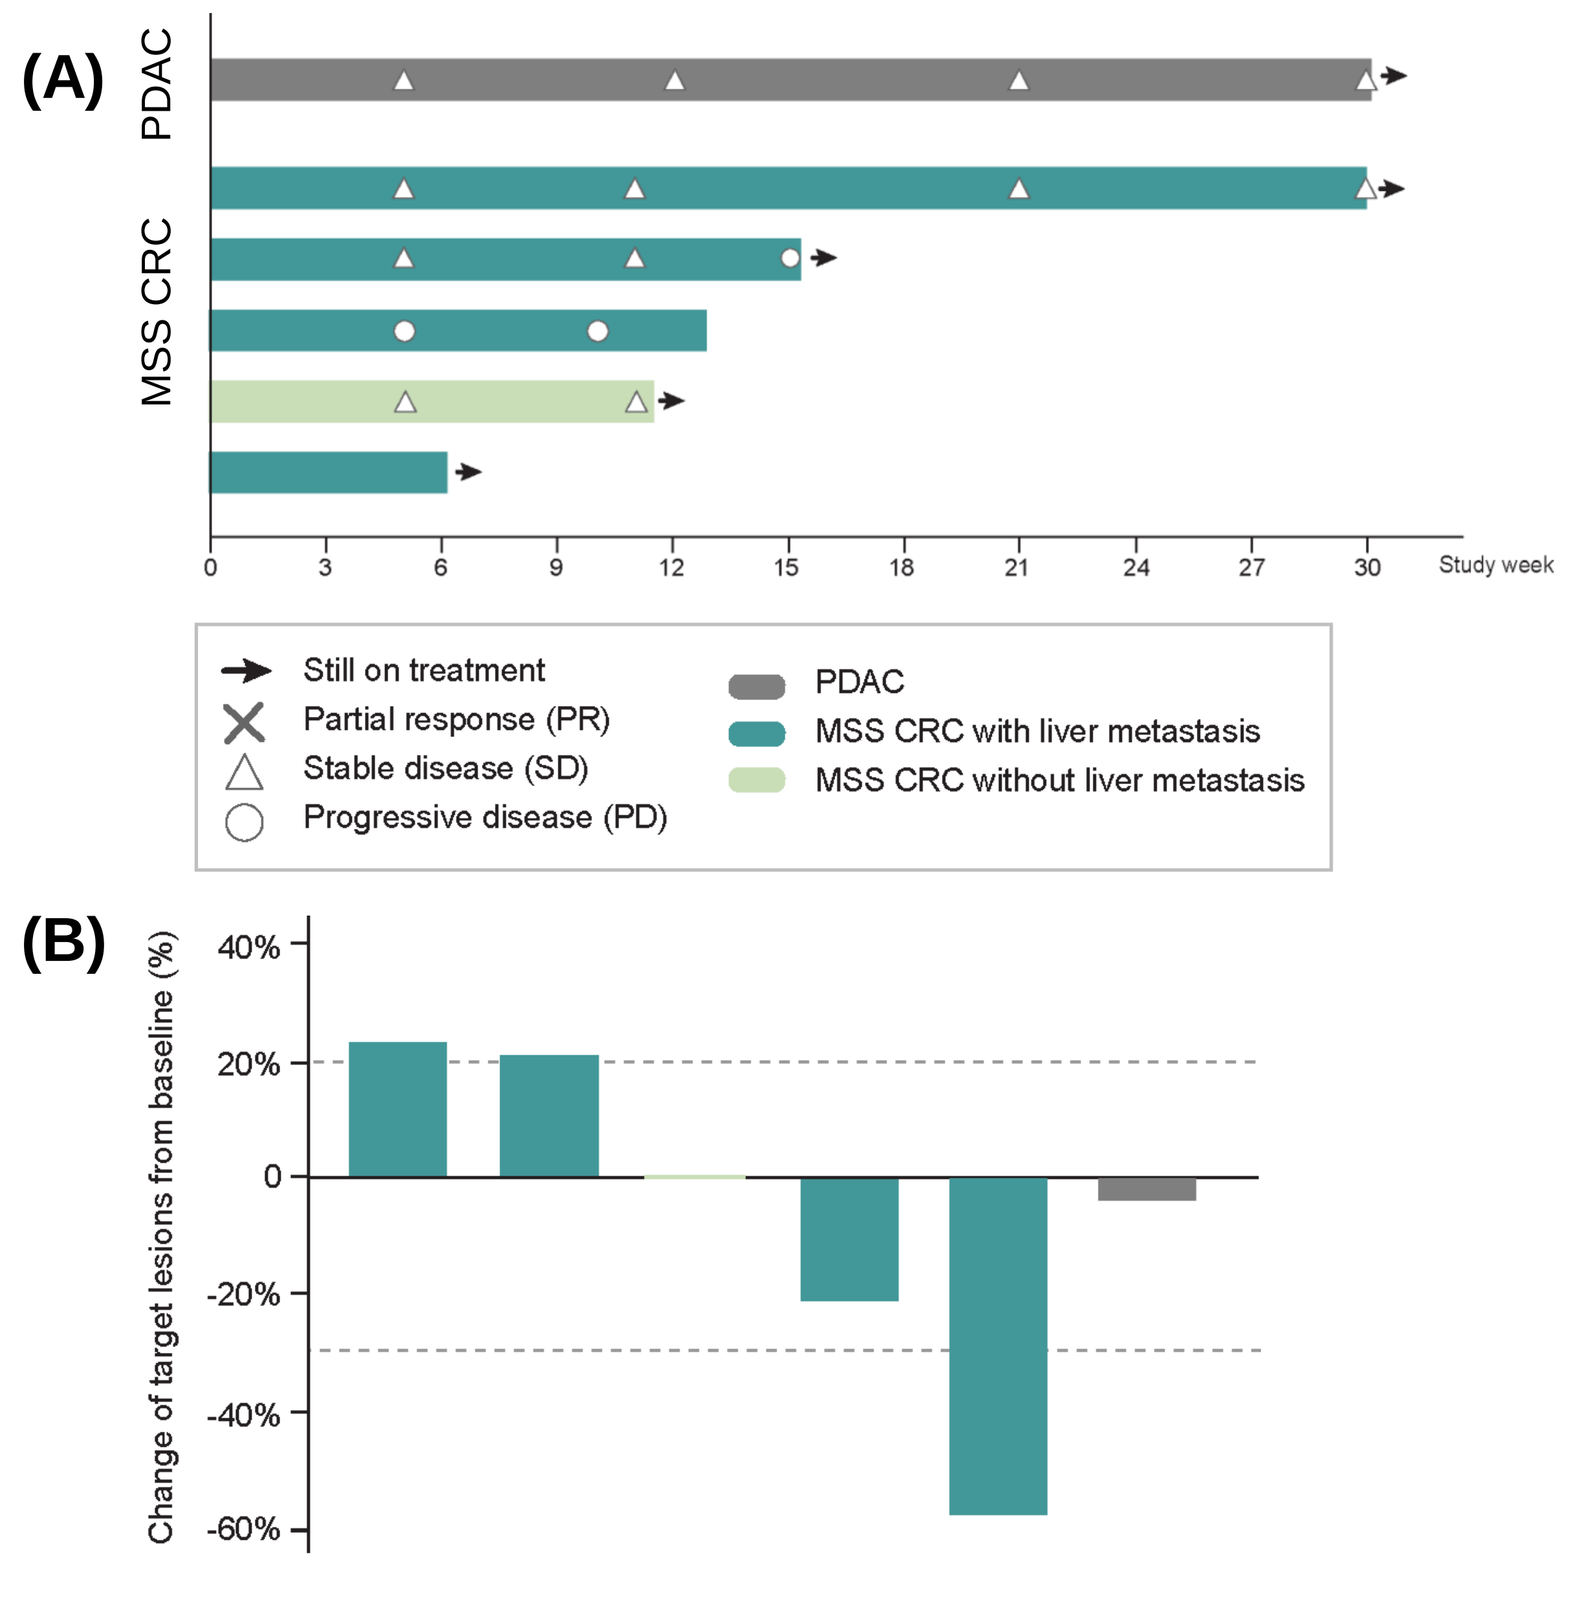
(A)
PDAC
MSS CRC
Study week
Still on treatment
Partial response (PR)
Stable disease (SD)
Progressive disease (PD)
PDAC
MSS CRC with liver metastasis
MSS CRC without liver metastasis
(B)
Change of target lesions from baseline (%)

Figure 7. Response to ADG126 + TORI in gastrointestinal epithelial "cold tumors" including MSS CRC and PDAC (A) Swimmer plot (B) Waterfall plot

This study is funded by Adagene Inc. Contact ir@adagene.com. Sincere appreciation for all patients, their families, investigators and staff. The presenting author Dr. Zha is employed by Adagene Inc. We would like to acknowledge Dr. Xuesong Chen annd Dr. Wenqing Song (clinical PK assay development for ADG126), Ms Carissa Lim (graphic design) and Dr. Joe Yeong's lab at IMCB, A*STAR (the immunofluorescence analysis on the tumor biopsy specimens).
Copies of this poster obtained through the QR code are for personal use only and may not be reproduced without written permission of the authors