2020 AACR Annual Meeting; Abstract Number 4538

A Safe and Potent Agonist ADG106 Targeting a Unique Epitope of CD137 with Novel Mechanism of Actions

Authors: Guizhong Liu, Zhengxi Dai, Jiagui Qu, Xiaohong She, Yan Li, Felix Du, Peter Luo
Adagene Inc, Suzhou, CHINA

Abstract

- Antagonist antibodies targeting immune checkpoint co-inhibitory receptor such as PD-1/PD-L1 have achieved great clinical success in cancer immunotherapy. However, agonistic antibodies targeting the co-stimulatory T cell receptors in the TNFR superfamily, and CD137 in particular, has suffered significant setback due to either dose-dependent severe liver toxicity by Urelumab, or modest clinical anti-tumor activity by Utomilumab[1]. ADG106, a safe and potent antibody targeting a unique epitope of CD137, is shown to hold great promise to achieve better efficacy and safety profile for immunotherapy in preclinical and phase I studies.
- ADG106, a fully human agonistic anti-CD137 IgG4 mAb, binds to a unique and conserved epitope with multiple cross-species reactivity, and overlaps with ligand binding site. This unique antibody acts in a novel mechanism with the following attributes: 1) it agonizes CD137 in a natural ligand-like fashion; 2) it blocks ligand to disable its reverse signaling; 3) it exhibits strong but specific Fc receptor mediated crosslinking in comparison with Utomilumab. ADG106 shows robust dose-dependent single agent anti-tumor activity in multiple syngeneic mouse tumor models and induces durable antigen-specific memory immunity that protects animals from the same re-challenged tumor cells.
- Preclinical studies demonstrate that ADG106 enhances activation and inflammatory cytokine release of primed T cells alone or together with other immunomodulatory agents in vitro, as well as exhibits synergistic in vivo anti-tumor activity in combination with variety of other cancer therapies, including checkpoint inhibitors, chemotherapies and targeted agents, radiation, etc. Mechanistic analyses suggest that ADG106 stimulates tumor infiltration and expansion of CD4+ and CD8+ T cells, thereby promoting the antitumor responses.
- ADG106 is well-tolerated in rodents and monkeys, with NOAEL up to 100mg/kg weekly doses in rodents and 200 mg/kg weekly doses in monkeys. Liver toxicity has not been observed.
- These findings support that ADG106-boosted immune response could offer an effective but alternative solution for cancer immunotherapy in single and combination therapies, especially for non-responders to current PD-1/PD-L1 based immunotherapies.

High affinity binding to CD137 across species

CD137 binding affinity of ADG106 by SPR (Biacore)

	Human	Cyno Monkey	Rat	Mouse
KD (nM)	3.73	4.77	14.7	21.5

ADG106 exhibits broad-species cross-reactivity with CD137 of human, cynomolgus monkey, and rodent origins. The rodent CD137 cross-reactivity enables pharmacological evaluation of ADG106 directly using syngeneic mouse tumor models as single agent, as well as in combination with other cancer therapies.

ADG106 is a CD137 ligand blocking antibody

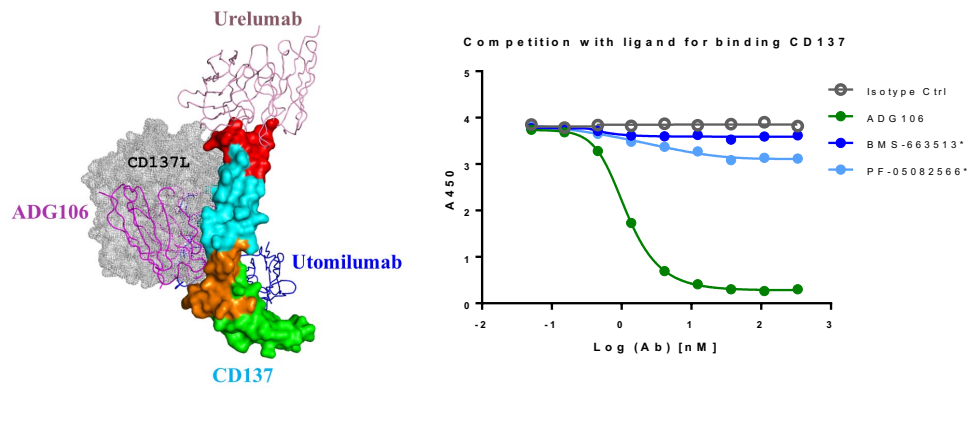
Urelumab
CD137L
ADG106
Utomilumab
CD137
Competition with ligand for binding CD137
Isotype Ctrl
ADG106
BMS-663513*
PF-05082566*
A450
Log (Ab) [nM]

Fig. 1. ADG106 overlaps with CD137 ligand for binding to CD137. The cartoon shows the relative binding sites on CD137 protein by 3 clinical anti-CD137 antibodies. Urelumab (BMS) binds to the N-terminus of CD137, Utomilumab (Pfizer) binds to the membrane-proximal region of CD137, whereas ADG106 overlaps with CD137 ligand binding site in the central region of CD137. An in vitro protein-protein interaction ELISA assay demonstrates that ADG106 is a potent CD137 ligand blocking antibody than the other two reference antibodies.

Contact information: peter_luo@adagene.com

ADG106 acts as a crosslinking-dependent CD137 agonist

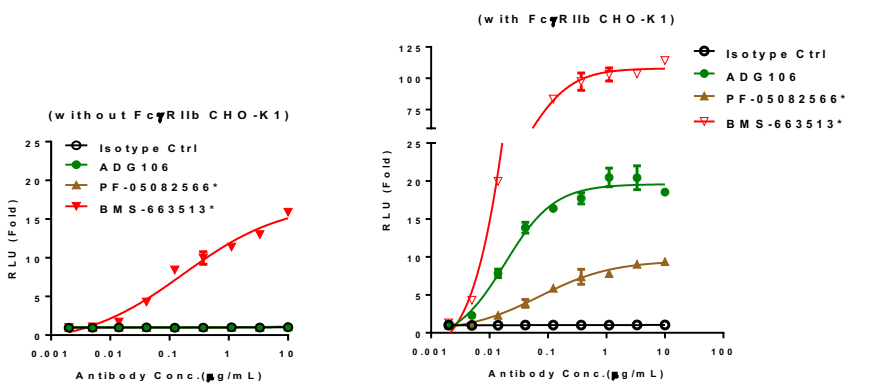
(without FcγRIIb CHO-K1)
(with FcγRIIb CHO-K1)
Isotype Ctrl
ADG106
PF-05082566*
BMS-663513*
RLU (Fold)
Antibody Conc.(μg/mL)

Fig. 2. Activation of CD137 receptor signaling. The CD137-expressing NFκB-Luc Jurkat reporter cells were stimulated with the anti-CD137 antibodies in the absence or presence of co-cultured CHO-K1 cells expressing human FcγRIIb. Luciferase activity was measured by bioluminescence assay. Results demonstrate that both ADG106 and PF-05082566 can activate CD137 cell signaling, only when crosslinked, such as by FcγRIIB, whereas BMS-663513 is able to activate CD137 cell signaling in dependent of crosslinking, In the presence of FcγRIIB-mediated crosslinking, BMS-663513 stimulates the highest CD137 signaling, and ADG106 exhibits stronger CD137 agonistic activity than PF-05082566.

ADG106 co-stimulates human T cell activation *in vitro*

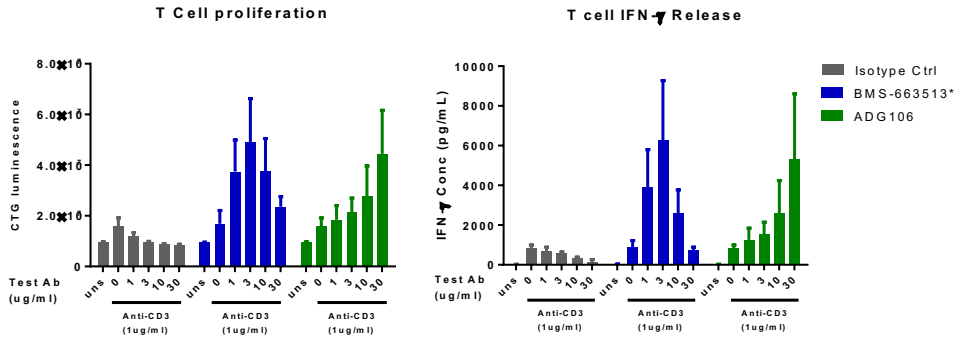
T Cell proliferation
T cell IFN-γ Release
CTG luminescence
IFN-γ Conc (pg/mL)
Test Ab (ug/ml)
Anti-CD3 (1ug/ml)
Isotype Ctrl
BMS-663513*
ADG106

Fig. 3. Anti-CD137 antibodies co-activate human T cells in vitro. Human T cells isolated from PBMC were cultured in 96-well microplates precoated with anti-CD3 (1 μg/ml) and various concentrations of anti-CD137 antibodies. T cell proliferation and IFN-g release were detected as readout of T cell activation. The results demonstrate that both BMS-663513 and ADG106 can enhance human T cell activation as co-activator.

In vivo antitumor efficacy of ADG106 as single agent

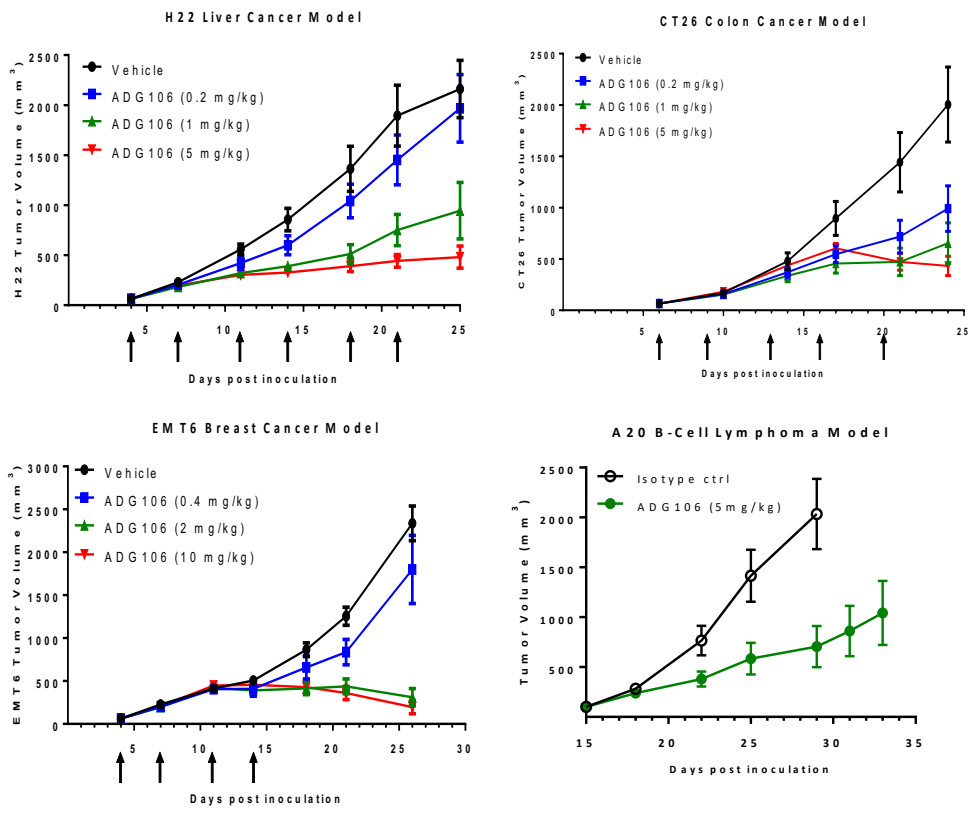
H22 Liver Cancer Model
Vehicle
ADG106 (0.2 mg/kg)
ADG106 (1 mg/kg)
ADG106 (5 mg/kg)
CT26 Colon Cancer Model
EMT6 Breast Cancer Model
ADG106 (0.4 mg/kg)
ADG106 (2 mg/kg)
ADG106 (10 mg/kg)
A20 B-Cell Lymphoma Model
Isotype ctrl
ADG106 (5mg/kg)
Tumor Volume (mm³)
Days post inoculation

Fig. 4. In vivo anti-tumor efficacy of ADG106 single agent in different mouse tumor models.

Induction of antitumor memory response

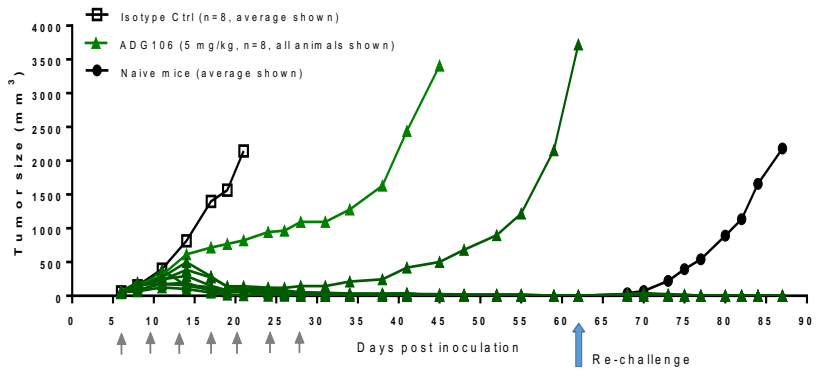
Isotype Ctrl (n=8, average shown)
ADG106 (5 mg/kg, n=8, all animals shown)
Naive mice (average shown)
Tumor size (mm³)
Days post inoculation
Re-challenge

Fig. 5. Development of antitumor memory response by ADG106 treatment in mouse H22 liver cancer model.

ADG106 synergizes with other anti-cancer therapies

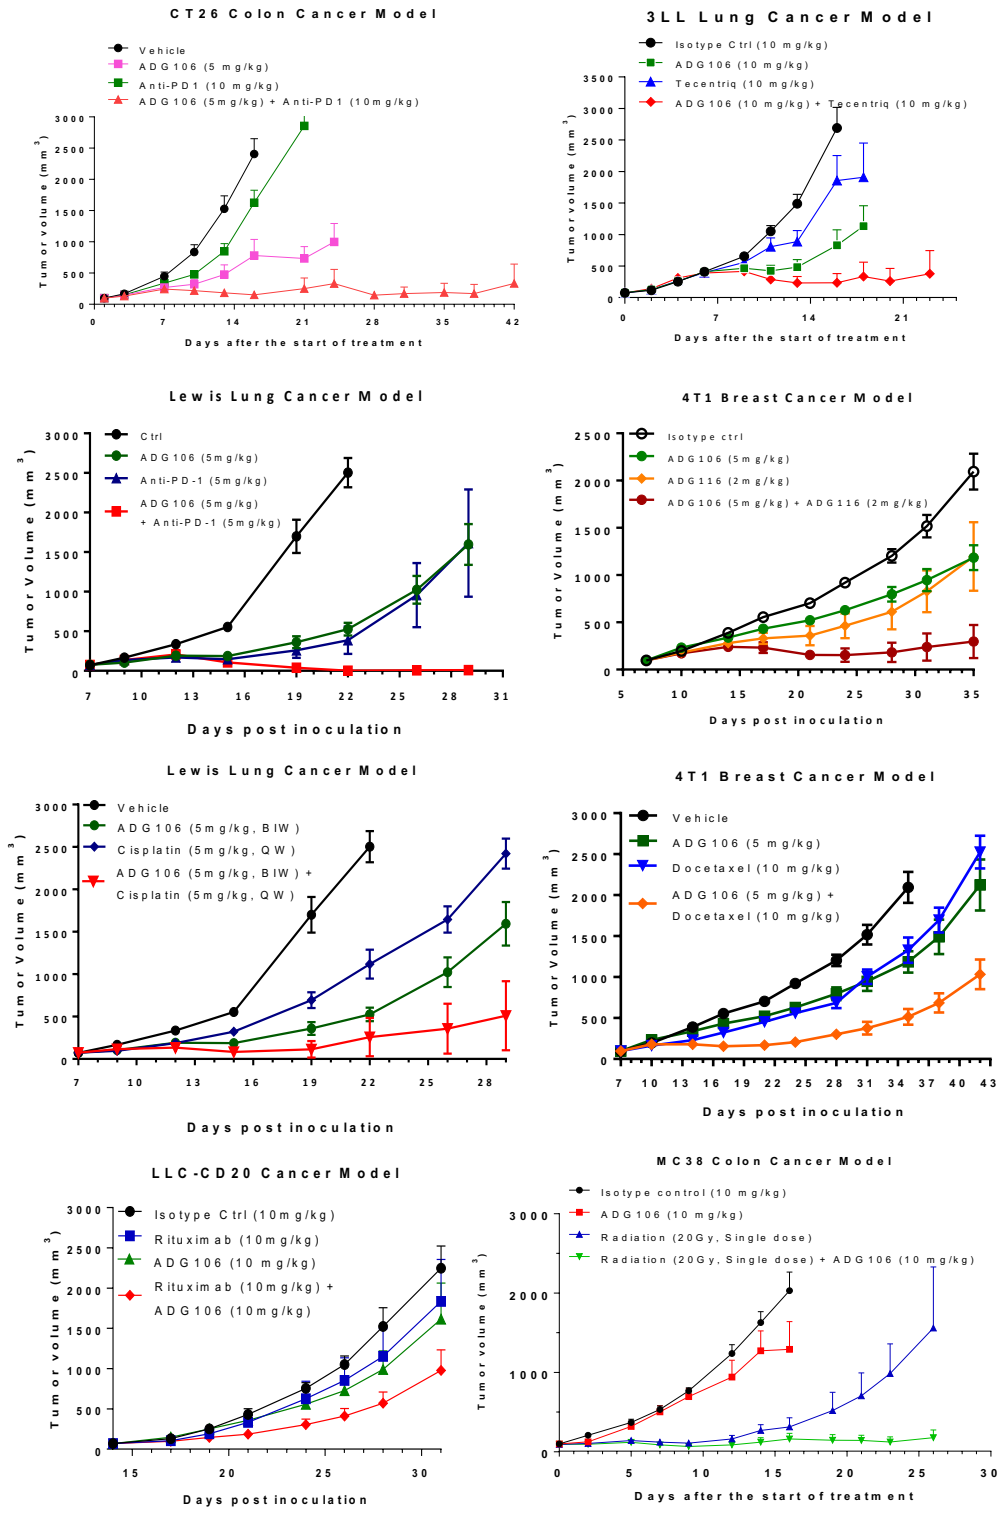
CT26 Colon Cancer Model
Vehicle
ADG106 (5 mg/kg)
Anti-PD1 (10 mg/kg)
ADG106 (5mg/kg) + Anti-PD1 (10mg/kg)
3LL Lung Cancer Model
Isotype Ctrl (10 mg/kg)
ADG106 (10 mg/kg)
Tecentriq (10 mg/kg)
ADG106 (10 mg/kg) + Tecentriq (10 mg/kg)
Lewis Lung Cancer Model
Ctrl
ADG106 (5mg/kg)
Anti-PD-1 (5mg/kg)
ADG106 (5mg/kg) + Anti-PD-1 (5mg/kg)
4T1 Breast Cancer Model
Isotype ctrl
ADG106 (5mg/kg)
ADG116 (2mg/kg)
ADG106 (5mg/kg) + ADG116 (2mg/kg)
Lewis Lung Cancer Model
Vehicle
ADG106 (5mg/kg, BIW)
Cisplatin (5mg/kg, QW)
ADG106 (5mg/kg, BIW) + Cisplatin (5mg/kg, QW)
4T1 Breast Cancer Model
Vehicle
ADG106 (5 mg/kg)
Docetaxel (10 mg/kg)
ADG106 (5 mg/kg) + Docetaxel (10 mg/kg)
LLC-CD20 Cancer Model
Isotype Ctrl (10mg/kg)
Rituximab (10mg/kg)
ADG106 (10 mg/kg)
Rituximab (10mg/kg) + ADG106 (10mg/kg)
MC38 Colon Cancer Model
Isotype control (10 mg/kg)
ADG106 (10 mg/kg)
Radiation (20Gy, Single dose)
Radiation (20Gy, Single dose) + ADG106 (10 mg/kg)
Tumor Volume (mm³)
Days post inoculation
Days after the start of treatment

Fig. 6. In vivo anti-tumor efficacy of ADG106 in combination with various cancer therapies, including checkpoint inhibitors anti-PD-1/anti-PD-L1, anti-CTLA-4 (ADG116), chemotherapy Cisplatin or Docetaxel, targeted therapy Rituximab, and Radiation. The results demonstrate that ADG106 can synergize with these various cancer therapies to enhance antitumor efficacy in different mouse syngeneic cancer models.

ADG106 stimulates tumor infiltration of T cells

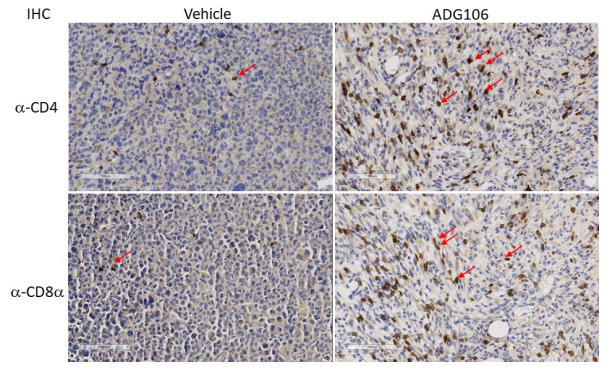
IHC
Vehicle
ADG106
α-CD4
α-CD8α

Fig. 7. Representative IHC images of mouse $CD4^+$ (upper panel) and $CD8^+$ (lower panel) T cells in mouse H22 tumors after treatment with vehicle control (left panel) or ADG106 (right panel). $CD4^+$ or $CD8^+$ T cells were stained in Brown, as indicated by the red arrow, in the background of nuclear counterstain by Hematoxylin. Results demonstrate that ADG106 treatment significantly increases tumor infiltrating T cells.

ADG106 is well tolerated in preclinical animal models

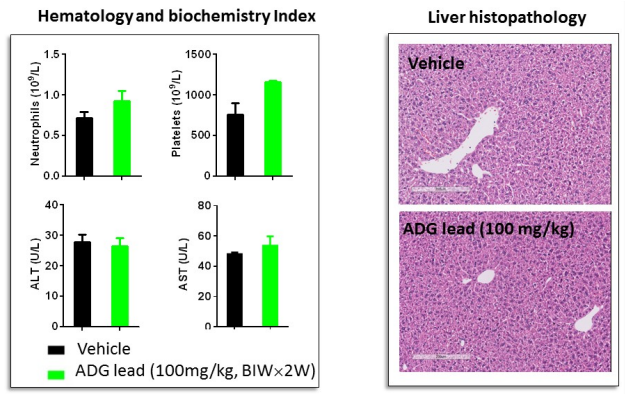
Hematology and biochemistry Index
Liver histopathology
Neutrophils (10⁹/L)
Platelets (10⁹/L)
ALT (U/L)
AST (U/L)
Vehicle
ADG lead (100mg/kg, BIW×2W)
Vehicle
ADG lead (100 mg/kg)

Fig. 8. ADG106 is well tolerated in mice. No toxicity was observed in mice treated with ADG106 at 100 mg/kg twice a week for 2 weeks. There were no changes in hematology parameters and ALT/AST liver enzyme increase. Liver histopathology also appeared normal, indicating the absence of liver toxicity after ADG106 treatment. In addition, the GLP 29-day weekly repeat dose toxicology studies indicate NOAEL ≥ 100 mg/kg in rats and ≥200 mg/kg in cynomolgus monkeys.

Summary

- ADG106 binds to a unique epitope of CD137 across a broad species .
- ADG106 acts with dual mechanisms: 1) directly activates CD137 receptor signaling, and 2) block CD137 ligand mediated reverse signaling.
- ADG106 activity is dependent on FcγR mediated crosslinking, and superior to Utomilumab.
- ADG106 has potent antitumor activity in mouse tumor models as single agent, as well as in combination with various anticancer therapies, including checkpoint inhibitors such as anti-PD-1/PD-L1, anti-CTLA-4, chemotherapies, targeted agent, as well as radiation.
- ADG106 can induce long-term protective memory responses.
- In vitro, ADG106 can co-activate T cells and stimulate IFN-γ release; in vivo, ADG106 can stimulate tumor infiltrating T cells, which mediates the antitumor response.
- GLP toxicity studies demonstrate that ADG106 is well tolerated in Rats (NOAEL≥100 mg/kg) and Cynomolgus monkeys (NOAEL≥200 mg/kg).

References

1. Chester et al. Immunotherapy targeting 4-1BB: mechanistic rationale, clinical results, and future strategies. *Blood* 2018, 131: 49

Acknowledgement: We are grateful to colleagues who provided support on this project.